Pharmacokinetics of RLS-0071, a Novel Anti-Inflammatory Peptide, in Newborns With Moderate or Severe Hypoxic Ischemic Encephalopathy

Kenji M. Cunnion, MD, MPH[1,2,3,4], Neel K. Krishna PhD[1], Zachary A. Vesoulis, MD MSCI[5]

[1]ReAlta Life Sciences, Norfolk, Virginia, USA; [2]Department of Pediatrics, Eastern Virginia Medical School, Norfolk, Virginia, USA; [3]Children's Hospital of the King's Daughters, Norfolk, Virginia, USA; [4]Children's Specialty Group, Norfolk, Virginia, USA; [5]Department of Pediatrics, Washington University School of Medicine, St. Louis, Missouri, USA

Background

- Hypoxic ischemic encephalopathy (HIE) is a common cause of infant mortality and life-long neurocognitive disabilities with no currently approved pharmacological treatments available
- RLS-0071 (pegtarazimod) is a novel anti-inflammatory peptide that inhibits complement activation at C1, as well as myeloperoxidase activity and neutrophil extracellular trap formation
- Prior pharmacokinetic (PK) analysis of RLS-0071 in animal and adult human studies demonstrated a two-compartment model with a bi-exponential decline in plasma concentration driven by rapid distribution into tissues
- RLS-0071 is currently being evaluated in the STAR study, a phase 2 randomized placebo-controlled dose-escalation trial in neonates with moderate or severe HIE (NCT05778188)

Objective

To characterize the pharmacokinetics (PK) profile of RLS-0071 in newborns with HIE to assess safety

Methods

Sample Collection and RLS-0071 Detection:

- Participating infants in the STAR study were dosed within 10 hours of birth. RLS-0071 was administered at 3 mg/kg IV (first dosing cohort) and continued every 8 hours for a total of 10 doses
- Blood samples were obtained pre-dose, at the end of first infusion (Cmax), 1 hour post dose (close to the inflection point in the bi-exponential kinetics) and at 8 hours post dose (Cmin)
- RLS-0071 plasma concentration was measured by liquid chromatography/mass spectrometry (LC/MS)

Modeling and Simulation of PK curves:

- Neonatal PK modeling was performed using a two-compartment model and results compared against adult PK profiles derived from the RLS-0071 healthy volunteer Phase 1 trial (NCT05298787)
- To simulate a pediatric population, an adult model was adapted with 30% variability in clearance (CL) and volume of distribution (Vd)
- This model was then applied to neonates weighing 3 kg, with 100 simulations performed by sampling from the ETA distributions of the PK parameters. Simulations were conducted using allometric scaling and assumed a conservative 50% reduction in CL to account for delayed organ maturation and hypothermia

Results

- RLS-0071 plasma concentrations for 10 HIE neonates (8 moderate, 2 severe) following the first dose and multiple doses at 3 mg/kg are summarized in Table 1
- No drug accumulation was observed, and concentrations were within established safe plasma levels in adults
- RLS-0071 PK profile in neonates follows a two-compartment model with a bi-exponential curve consistent with adult kinetics
- HIE infant PK curves for RLS-0071 at the 3 mg/kg/dose were similar to adult human curves at the 2 mg/kg/dose (Figure 1)

Table 1. PK plasma values for RLS-0071 at 3 mg/kg IV Q8hr

	C_{max} (min, max) mcg/ml	C_{min} (min, max) mcg/ml	$AUC_{0\text{-}8hr}$ (min, max) mcg/ml/hr
First Dose	11.7 (1.9, 72.80)	ND	ND
Multiple Doses	10.2 (1.1, 26.7)	0.05 (0.02, 0.10)	7.3 (1.0, 16.5)

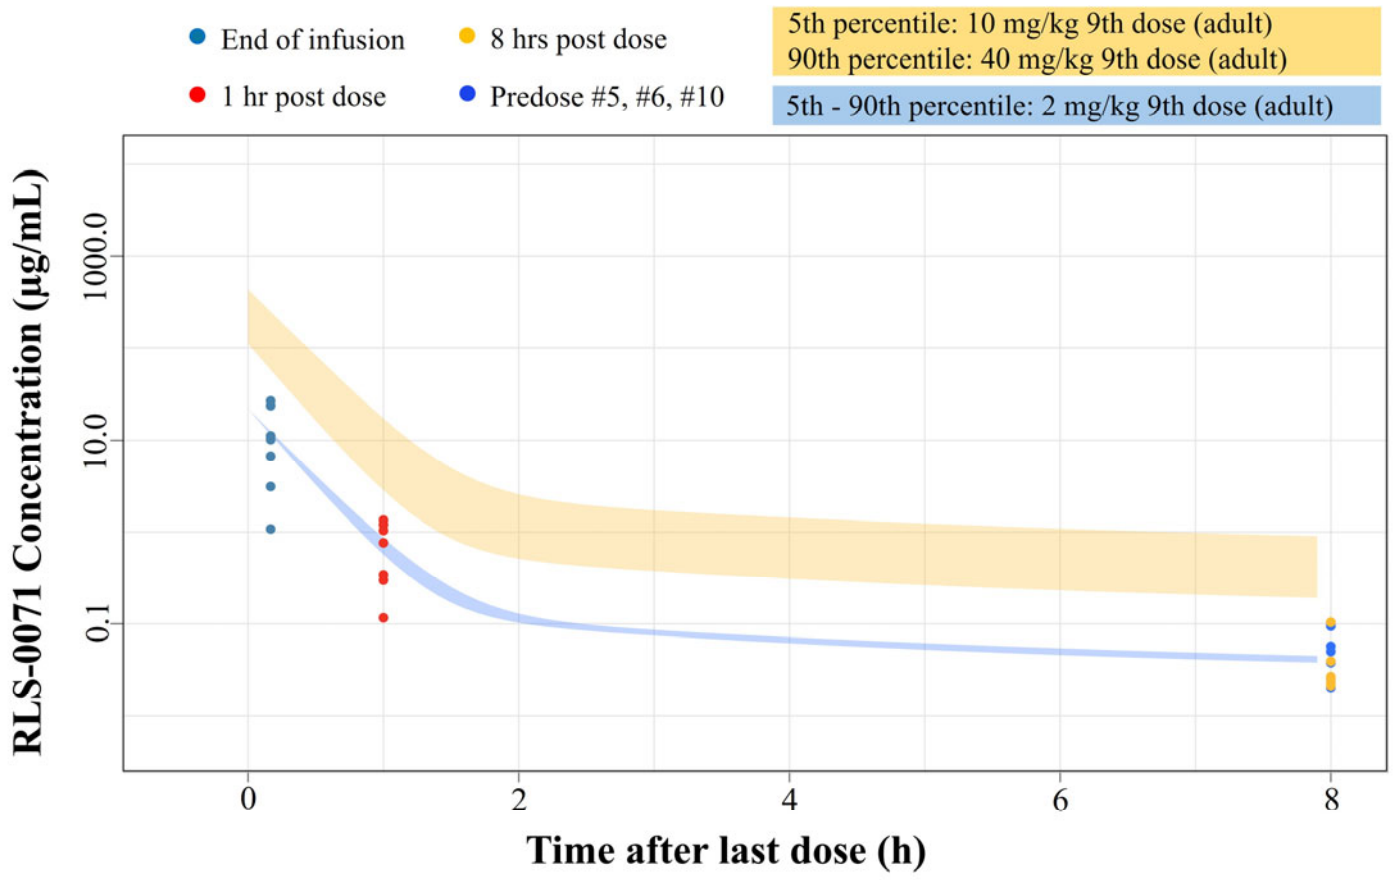
End of infusion
8 hrs post dose
1 hr post dose
Predose #5, #6, #10
5th percentile: 10 mg/kg 9th dose (adult)
90th percentile: 40 mg/kg 9th dose (adult)
5th - 90th percentile: 2 mg/kg 9th dose (adult)
RLS-0071 Concentration (μg/mL)
1000.0
10.0
0.1
0
2
4
6
8
Time after last dose (h)

Figure 1. Comparison of pediatric and adult RLS-0071 PK curves after multiple doses. Yellow and blue shaded ribbons represent adult post-hoc simulations. Colored dots represent pediatric data at 3 mg/kg after 10th dose. Predose #5, #6, #10 are plotted at 8 h after 10th dose.

Results

- Simulation results for a single dose and multiple doses at assumed clearance levels are summarized in Table 2
- Assuming a 50% reduction in CL, simulation results predicted that 30% of infants receiving a 3 mg/kg dose of RLS-0071 would reach the target peak concentration (Cmax = 17 mcg/ml), associated with optimal efficacy in the animal model

Table 2. Predicted proportion of participants above threshold of 17 mcg/ml.

CL reduction	Dose (mg/kg)	N_{dose}	n	% above threshold
50% reduction	3	1	100	30
50% reduction	3	10	100	30
50% reduction	10	1	100	99
50% reduction	10	10	100	99
No reduction	3	1	100	1
No reduction	3	10	100	1
No reduction	10	1	100	83
No reduction	10	10	100	83

Conclusions

- The pharmacokinetics of RLS-0071 in HIE neonates followed a bi-exponential profile and was consistent with PK curves in adults
- Following multiple 3 mg/kg IV doses, RLS-0071 plasma levels in neonates were within previously established safety limits
- These results establish a safety and PK profile in HIE neonates that is reassuring for the continued investigation of RLS-0071 in HIE clinical trials

Disclosures

Funding for this research was provided by ReAlta Life Sciences. N. Krishna and K. Cunnion are employees of ReAlta Life Sciences and receive salary support from ReAlta

Acknowledgements

We would like to acknowledge Certara for use of their modeling and simulation services